Safety and Tolerability of a Novel Tissue Factor / Factor VIIa Inhibitor rNAPc2 in Patients with NSTE-ACS Managed Invasively: Results from the ANTHEM-TIMI 32 Trial

Richard G. Bach, Robert P. Giugliano, Daniel I. Simon, Marc J. Schweiger, Massoud A. Leesar, Stephen D. Wiviott, Michael A. Goulder, Steven R. Deitcher, Eugene Braunwald; Washington University Medical Center, St. Louis, MO, Brigham and Women's Hospital, Boston, MA

INTRODUCTION

Despite intensive antiplatelet, anticoagulant, and anti-ischemic therapy, and early invasive management, recurrent MI or death remain frequent events among high-risk patients with non-ST-elevation acute coronary syndrome (nSTE-ACS). In patients with ACS, plaque rupture and PCI expose **tissue factor** that can rapidly generate thrombin and activate coagulation. Current standard antithrombotic therapies (heparin, aspirin, clopidogrel, glycoprotein (GP) IIb/IIIa blockers) **do not adequately suppress thrombin generation.**

Recombinant nematode anticoagulant protein c2 (**rNAPc2**) is an 85-amino acid serine protease inhibitor originally isolated from the hookworm parasite *Ancylostoma caninum*. The anticoagulant activity results from **inhibition of the catalytic complex of factor VIIa/tissue factor (fVIIa/TF)** by a unique mechanism. The elimination half-life is estimated to be 50-60 hours in man. Prior studies of rNAPc2 have demonstrated its novel potential to ***inhibit thrombin generation*** in patients undergoing ***elective*** PCI.

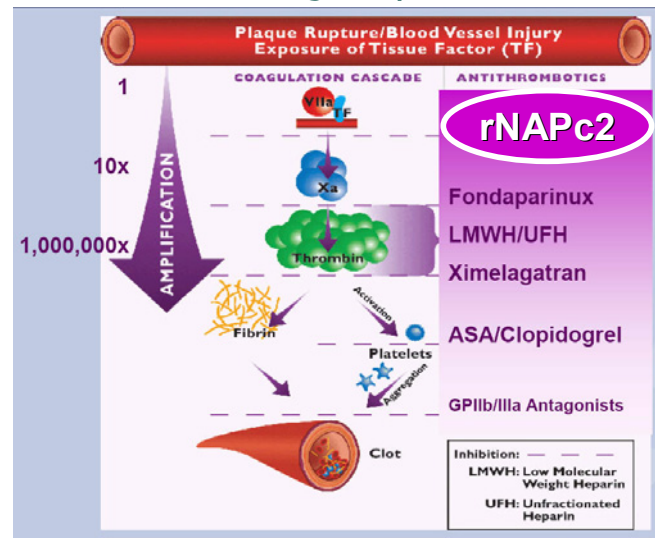
Plaque Rupture/Blood Vessel Injury
Exposure of Tissue Factor (TF)
COAGULATION CASCADE
ANTITHROMBOTICS
1
10x
1,000,000x
AMPLIFICATION
VIIa TF
Xa
Thrombin
Fibrin
Platelets
Clot
rNAPc2
Fondaparinux
LMWH/UFH
Ximelagatran
ASA/Clopidogrel
GPIIb/IIIa Antagonists
Inhibition:
LMWH: Low Molecular Weight Heparin
UFH: Unfractionated Heparin

STUDY PURPOSE

The goal of ANTHEM-TIMI 32 was to evaluate safety (and efficacy) of a range of doses of rNAPc2 in patients with nSTE-ACS managed with standard therapies including early invasive strategy.

STUDY DESIGN:

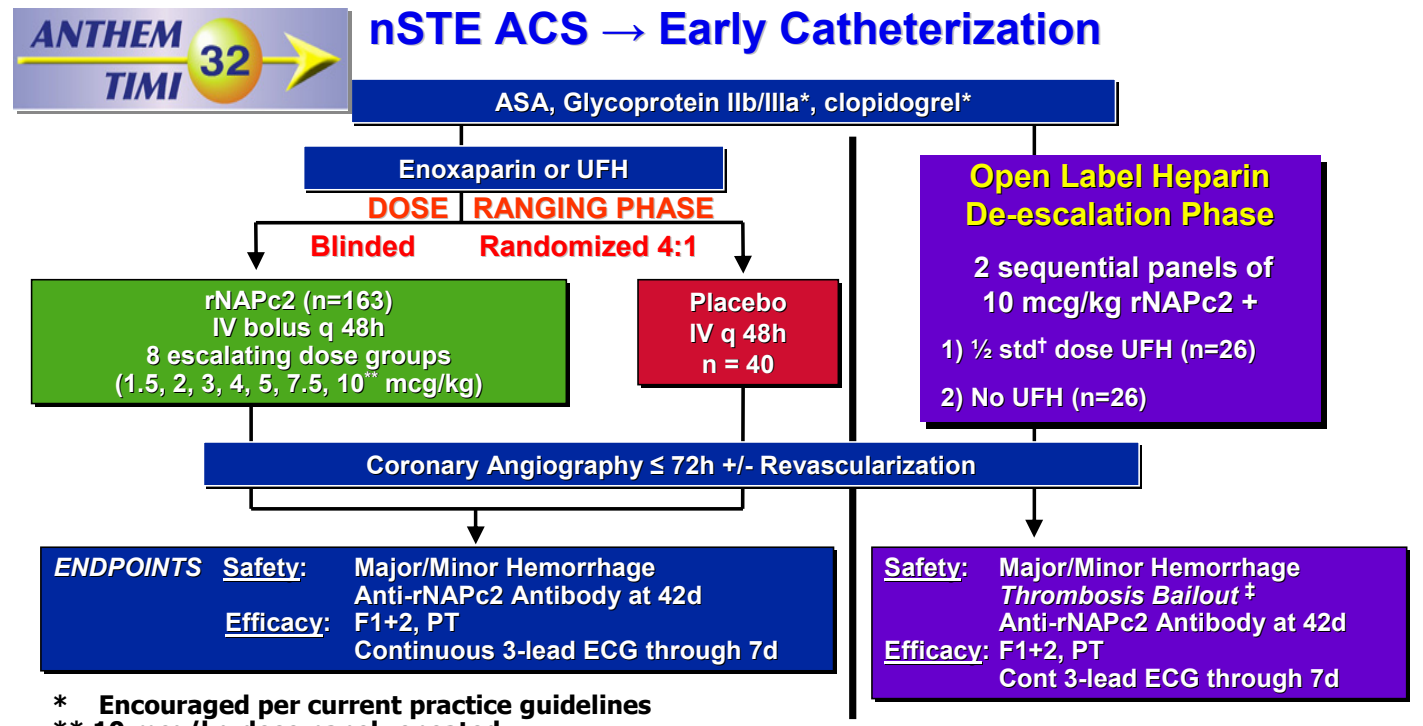
ANTHEM TIMI 32
nSTE ACS → Early Catheterization
ASA, Glycoprotein IIb/IIIa*, clopidogrel*
Enoxaparin or UFH
DOSE RANGING PHASE
Blinded
Randomized 4:1
rNAPc2 (n=163)
IV bolus q 48h
8 escalating dose groups
(1.5, 2, 3, 4, 5, 7.5, 10** mcg/kg)
Placebo
IV q 48h
n = 40
Open Label Heparin De-escalation Phase
2 sequential panels of 10 mcg/kg rNAPc2 +
1) ½ std† dose UFH (n=26)
2) No UFH (n=26)
Coronary Angiography ≤ 72h +/- Revascularization
ENDPOINTS Safety: Major/Minor Hemorrhage
Anti-rNAPc2 Antibody at 42d
Efficacy: F1+2, PT
Continuous 3-lead ECG through 7d
Safety: Major/Minor Hemorrhage
Thrombosis Bailout ‡
Anti-rNAPc2 Antibody at 42d
Efficacy: F1+2, PT
Cont 3-lead ECG through 7d

* Encouraged per current practice guidelines
** 10 mcg/kg dose panel repeated
† 30 U/kg bolus (max 2500 U); 6 U/kg/h (max 600 U/h)
‡ Heparin de-escalation phase ONLY and Investigator reported; defined as use of open-label anticoagulant to manage thrombotic complication

Note: The incidence of procedure-related thrombosis was prospectively collected in the open label heparin de-escalation phase only, and not among the 203 patients in the dose-ranging phase of the study.

ENTRY CRITERIA AND STUDY SCHEMA:

Inclusion:	Age 18-75, rest sx ≥ 5min w/i 48h c/w ACS
	+ markers or ST deviation or TIMI Risk Score ≥ 3
	Planned early invasive strategy
Exclusions:	STEMI, creatinine > 4, ↑bleed risk, planned CABG <7d

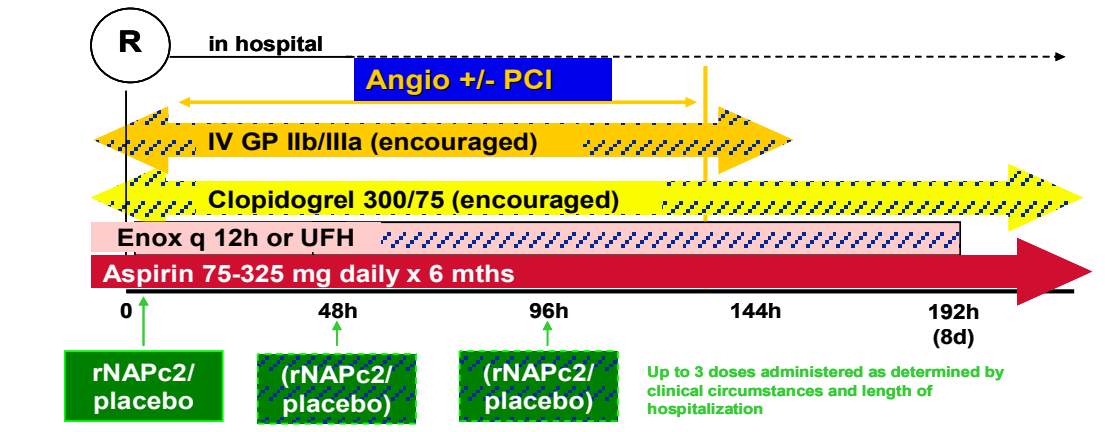
R
in hospital
Angio +/- PCI
IV GP IIb/IIIa (encouraged)
Clopidogrel 300/75 (encouraged)
Enox q 12h or UFH
Aspirin 75-325 mg daily x 6 mths
0
48h
96h
144h
192h (8d)
rNAPc2/ placebo
(rNAPc2/ placebo)
(rNAPc2/ placebo)
Up to 3 doses administered as determined by clinical circumstances and length of hospitalization

RESULTS

Table 1. Baseline Characteristics and Treatments

	rNAPc2 n = 215	placebo n = 40	P-value
Age (median), yrs	58	56	NS
Age ≥ 65 yrs, %	28	18	NS
Women,%	34	20	NS
nSTE-MI,%	54	50	NS
STdev>0.5mm,%	47	45	NS
Diabetes,%	34	33	NS
Mean TIMI Risk Score	3.7	4.0	NS
#Study Drug Doses 1/2/3,%	83/18/2	83/18/0	NS
*Enoxaparin/UFH,%	71/33	75/33	NS
Clopidogrel,%	81	85	NS
GP IIb/IIIa Inhibitor,%	54	53	NS
PCI/CABG/Medical,%	48/11/41	60/3/37	NS

*Excludes 52 patients in the heparin de-escalation phase

Figure 1: Higher Dose rNAPc2 Reduced:

(A) New Thrombin Generation

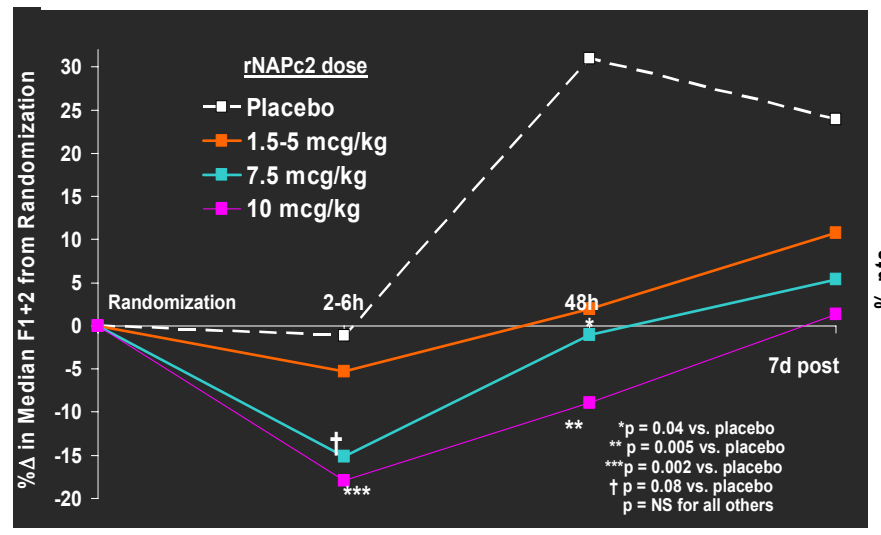
rNAPc2 dose
Placebo
1.5-5 mcg/kg
7.5 mcg/kg
10 mcg/kg
%Δ in Median F1+2 from Randomization
Randomization
2-6h
48h
7d post
*p = 0.04 vs. placebo
** p = 0.005 vs. placebo
***p = 0.002 vs. placebo
† p = 0.08 vs. placebo
p = NS for all others

(B) Ischemic Episodes

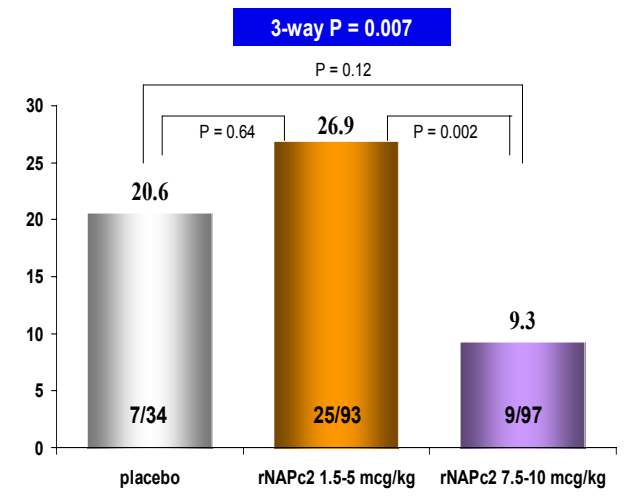
3-way P = 0.007
P = 0.12
P = 0.64
P = 0.002
% pts
20.6
26.9
9.3
7/34
25/93
9/97
placebo
rNAPc2 1.5-5 mcg/kg
rNAPc2 7.5-10 mcg/kg

Figure 1: (A) Change in F1+2, a marker of new thrombin generation, from baseline by treatment group and time; rNAPc2 at 10 and 7.5 mcg/kg significantly reduced new thrombin generation (B) Incidence of recurrent in-hospital ischemia by ST shifts on continuous ST segment monitoring was significantly reduced by rNAPc2 at 7.5-10 mcg/kg .

Table 2. Bleeding Outcomes, rNAPc2 vs. Placebo

	rNAPc2 n = 215	placebo n = 40	P-value
TIMI Major, n(%)	4(1.9)	0(0.0)	NS
Non-CABG TIMI Major	1(0.5)	0(0.0)	NS
TIMI Minor	4(1.9)	1(2.5)	NS
TIMI Major or Minor	8(3.7)	1(2.5)	NS
Any Reported Bleed*	28(13.0)	5(12.5)	NS
RBC Transfusion	14(6.5)	1(2.5)	NS
Ave # PRBC Transfused	2.2	2.0	NS
Fresh Frozen Plasma	2(0.9)	1(2.5)	NS
Thrombocytopenia ‡	3(1.4)	0(0.0)	NS
Platelet Transfusion	1(0.5)	1(2.5)	NS
Recombinant FVIIa	0	0	--
αrNAPc2 Ab+ @ 42d	23(13.0)†	0(0.0)	0.03

*Note: All major/minor bleeds were provoked by procedure/trauma as were 79% of the minimal bleeds
‡ Platelet counts of 99K, 98K, and 93K.
† % based on available data; No cases of allergic symptoms or anaphylaxis were reported

Figure 2: Adjudicated Bleeding by Anti-Thrombotic Medication Doses

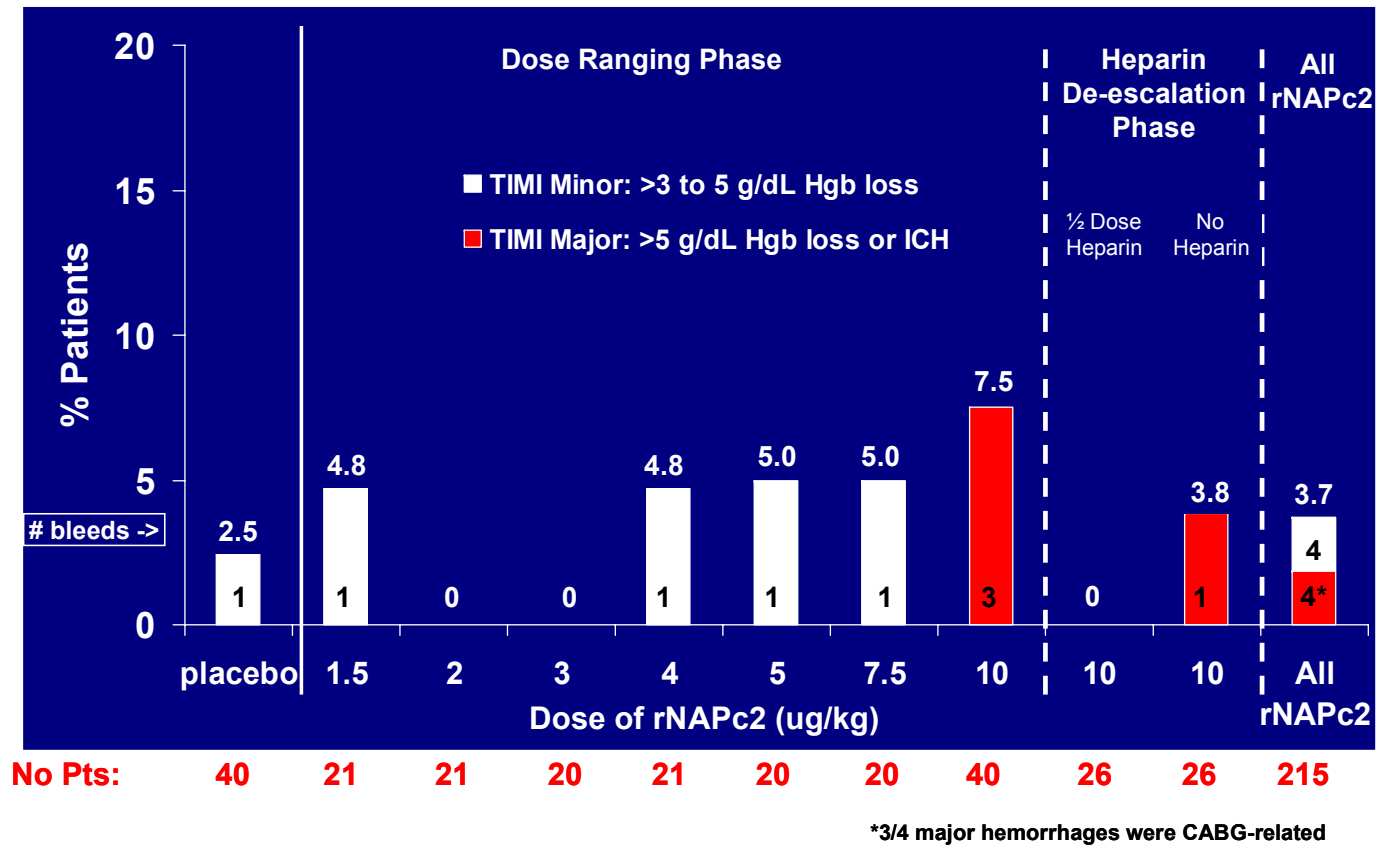
Dose Ranging Phase
Heparin De-escalation Phase
All rNAPc2
TIMI Minor: >3 to 5 g/dL Hgb loss
TIMI Major: >5 g/dL Hgb loss or ICH
½ Dose Heparin
No Heparin
% Patients
bleeds ->
2.5
4.8
4.8
5.0
5.0
7.5
3.8
3.7
4*
placebo
1.5
2
3
4
5
7.5
10
10
10
All rNAPc2
Dose of rNAPc2 (ug/kg)
No Pts: 40 21 21 20 21 20 20 40 26 26 215
*3/4 major hemorrhages were CABG-related

Table 3. Events According to Anti-Thrombotic Medication Doses

	rNAPc2 <10 mcg/kg n = 123	rNAPc2 10 mcg/kg+ Full dose hep n = 40	rNAPc2 10 mcg/kg+ ½ dose hep n = 26	rNAPc2 10 mcg/kg+ no heparin n = 26	placebo n = 40	P-value (5-way)
TIMI Major, n(%)	0(0.0)	3(7.5)	0(0.0)	1(3.8)	0(0.0)	0.011
Non-CABG TIMI Major	0(0.0)	1(2.5)	0(0.0)	0(0.0)	0(0.0)	NS
TIMI Minor	4(3.3)	0(0.0)	0(0.0)	0(0.0)	1(2.5)	NS
TIMI Major or Minor	4(3.3)	3(7.5)	0(0.0)	1(3.8)	1(2.5)	NS
Any Reported Bleed*	14(11.4)	8(20.0)	3(11.5)	3(11.5)	5(12.5)	NS
RBC Transfusion	6(4.9)	5(12.5)	0(0.0)	3(11.5)	1(2.5)	NS
Mean Peak PT	21.62±7.4	34.2±13.5	29.9±9.2	22.6±8.2	17.2±3.8	<0.0001
Procedure-related thrombosis‡	--	--	0	4(15)	--	0.051‡
αrNAPc2 Ab+ @ 42d†	11(11.2)	4(11.4)	5(20.8)	3(11.5)	0(0.0)	NS

*Note: All major/minor bleeds were provoked by procedure/trauma as were 79% of the minimal bleeds
‡Thrombosis prospectively collected only in open-label heparin de-escalation phase; p-value for "no heparin" vs. "½ dose" group; 1 additional pt in the "no heparin" group reported to have recurrent ischemia prior to cath that required bailout anticoagulation (UFH).
† % based on available data; No cases of allergic symptoms or anaphylaxis were reported

SUMMARY

- **rNAPc2 caused a dose-related increase in PT (Table 3); doses ≥7.5 mcg/kg were associated with a significant reduction of new thrombin generation at 2-6 and 48h (Figure 1A) and with a significant >50% reduction in the incidence of ischemic episodes by continuous ECG (Figure 1B).**
- **rNAPc2 was generally well tolerated; there were 8 significant hemorrhages (3.7%), including 4 major and 4 minor, among the 215 patients receiving rNAPc2 in the trial (p=NS vs. placebo, Table 2).**
- **All 4 major hemorrhages occurred at highest dose (10 mcg/kg) and were procedure-related (3/4 CABG-related, 1 cath access site). Shorter interval between the last dose of rNAPc2 and CABG was associated with a trend toward higher bleeding risk (40% vs 10% vs 0% for CABG performed <48h vs 48-96h vs. >96h, trend p=0.077). Bleeding was associated with CABG <96h after last dose of drug, but not anticoagulant type, use of clopidogrel, aspirin dose, or IIb/IIIa inhibitor use.**
- **Other safety outcomes, including rates of other (neither major nor minor) bleeding (9.8% vs 10%), frequency of transfusion (6.5% vs 2.5%), average # of units transfused (2.2 vs 2.0), and serious adverse events (30% vs 33%) were similar between rNAPc2 and placebo (all p = NS). No patients required treatment with activated FVII concentrate to reverse rNAPc2 anticoagulation.**
- **Anti-rNAPc2 antibody was detected at 42 days in 13% of patients receiving rNAPc2; no patient had an allergic reaction or anaphylaxis; there were no differences in clinical outcome or pharmacodynamic effects among patients who were + vs – for antibody.**
- **In the open-label heparin de-escalation phase, investigators reported 4(15%) procedure-related thromboses (and 1 episode of recurrent ischemia prior to cath) in the no heparin group and none with half-dose heparin.**

CONCLUSIONS:

1. **In NSTE-ACS patients managed invasively, TF targeted inhibition of thrombin generation by rNAPc2 reduced ischemia without a significant increase in non-CABG bleeding.**
2. **rNAPc2 was associated with a moderate incidence (13%) of anti-rNAPc2 antibody at 42 days, not associated with clinical sequelae.**
3. **Adjunctive heparin or inhibitors of the terminal portion of the coagulation cascade at PCI may reduce the risk of procedural thrombosis.**
4. **These data suggest favorable safety of rNAPc2, requiring confirmation in larger scale clinical trials.**