Statin-Like Dose-Dependent Reductions in LDL-C and Apolipoprotein B with ISIS 301012 - An Antisense Inhibitor of ApoB in Subjects with Polygenic Hypercholesterolemia -

Evan Stein,* JoAnn Flaim,† Emil Chuang,† Mark Wedel,† John Su,† Richard Geary,† Rosie Yu,† John Kastelein‡

*Metabolic & Atherosclerosis Research Center, Cincinnati OH, †Isis Pharmaceuticals, Inc., Carlsbad, CA; ‡Amsterdam Medical Center, University of Amsterdam, the Netherlands

Data Pending

400 mg/wk data will be included on completed poster for presentation at the meeting.

Abstract

Background: ISIS 301012 selectively inhibits apoB protein synthesis in the liver. Previously, we have shown an LDL-C reduction up to 70% in healthy volunteers in Phase I dose ranging studies with short term dosing. The safety and efficacy profile allowed for longer treatment duration (13 weeks), less frequent dosing (every other week), and a target population of polygenic hypercholesterolemics failing to meet target with diet and exercise alone.

Methods: Subjects (n=10 per group) with elevated LDL-C > 130 mg/dL were enrolled into 1 of 5 groups. The first 2 groups received four 200 mg doses of ISIS 301012 in the first 11 days (Days 1, 4, 8, and 11; "Load") followed by 100 mg qow (avg 50 mg/week), or 200 mg qow (avg 100 mg/week). The last 3 groups received 200, 300, or 400 mg every week without a load. Treatment to placebo was 4:1 in each cohort. Study drug was administered subcutaneously.

Results: The study is ongoing. A 12, 22, and 42% median reduction in LDL-C and a 22, 23, and 47% median reduction in apoB were observed in the 50, 100, and 200 mg/week dose groups, respectively, with negligible changes in the pooled placebo group. Preliminary results in the 300 mg/wk cohort showed a 42 and 41% reduction in LDL-C and apoB, respectively following the first 7 weeks of 13 weeks treatment. The most common adverse event has been mild, painless erythema at injection sites. No drug-related serious adverse events occurred. The drug was well tolerated. One subject displayed an isolated ALT >3x ULN 80 days after last dose.

Conclusions: The initial results show that ISIS 301012 achieves statin-like LDL-C reductions via a complementary mechanism to statins. Furthermore, achieving such LDL-C reductions with a dosing interval of every other week is feasible. Dose escalation to the 400 mg/wk dose group is underway.

Background

Apolipoprotein B & Atherogenic Lipoproteins

- ApoB is an essential component of the principal atherogenic lipoproteins VLDL, IDL, LDL, and Lp(a).
- LDL-C is a risk factor of CVD.
 - LDL-C lowering by statins reduces CVD event rate (TNT, IDEAL, PROVE-IT).
 - Aggressive LDL-C lowering by statins results in regression of atherosclerosis (REVERSAL, ASTEROID).
- Despite statin therapy, 70% of CV events are not prevented.[1]
 - Thus, there is a need for new agents that are complementary in mechanism and additive in effect to statins, and that exert beneficial effects on other risk markers, *e.g.* elevated triglycerides.[2]

ISIS 301012

- ISIS 301012 is a 2nd generation antisense oligonucleotide[3] complementary to the coding region of the human apoB mRNA.

Selective hybridization (binding) of ISIS 301012 to the cognate mRNA by Watson and Crick base-pairing results in RNase H-mediated degradation of the mRNA to prevent translation of the apoB protein. ApoB is the principal apolipoprotein of LDL and its metabolic precursor, VLDL.

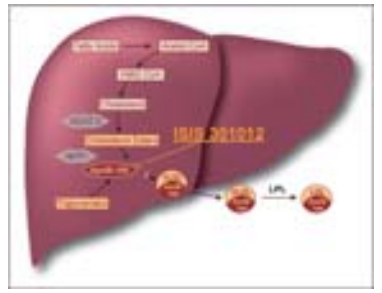
ISIS 301012

- The effects of ISIS 301012 at doses of 50 to 400 mg in healthy volunteers with primary hypercholesterolemia have recently been published.[4]
- The current study addresses the effects of ISIS 301012 mono-therapy in hypercholesterolemic subjects for 13 weeks. Other results presented at this meeting address the effects of ISIS 301012 in combination with statins in subjects of similar disposition.[5]

Study Design

- This Phase 2 clinical study was designed to examine the dose response of ISIS 301012 as a single agent in subjects with high cholesterol and unable to reach target by lifestyle changes alone.
- Fifty subjects were enrolled in one of five ISIS 301012 dose cohorts (4:1 ratio of active:placebo) at a single site in Germany.
- Study drug was administered by s.c. injection at a dose of 200 mg twice weekly for two weeks followed by 100 (50 mg/wk group) or 200 mg (100 mg/wk group) every other week for 11 weeks; and 200, 300, or 400 mg once per week for 13 weeks (200, 300, or 400 mg/wk groups) without an initial loading dose.
- The primary endpoint was percent reduction in LDL-C from baseline 2 weeks after a 3-month treatment period (Day 99).
- Safety and tolerability were assessed by the incidence, severity, and relationship of adverse events by treatment group. Follow-up period was for 6 months following the last dose.
- Secondary endpoints included % change from baseline in apoB, VLDL-C, HDL-C, non-HDL, total cholesterol, triglycerides, and the apoB/apo-A1 ratio.
- Exact Wilcoxon rank-sum tests were used to determine significant differences between placebo and the ISIS 301012-treated groups.

Baseline Characteristics

Characteristic	Placebo	50 mg/wk	100 mg/wk	200 mg/wk	300 mg/wk	400 mg/wk
n	10	8	8	8	8	8
Age	53	50	40	48	55	58
M/F	8/2	7/1	7/1	7/1	8/0	8/0
ApoB	130 (110-154)	156 (106-184)	129 (100-185)	130 (93-150)	139 (109-160)	150 (118-172)
LDL-C	162 (127-181)	172 (132-218)	154 (132-266)	173 (135-210)	178 (119-232)	206 (140-249)
Non-HDL-C	188 (156-238)	212 (158-250)	188 (152-276)	196 (150-225)	199 (153-239)	225 (163-260)
VLDL-C	23 (9-57)	17 (10-57)	19 (7-34)	17 (8-34)	23 (5-40)	30 (14-52)
HDL-C	54 (39-61)	55 (45-63)	57 (50-65)	54 (49-60)	50 (40-59)	52 (39-62)
Apo A1	134 (116-159)	137 (121-171)	142 (115-159)	127 (114-141)	114 (92-138)	128 (86-149)
Apo B:A1	1.0 (0.8-1.1)	1.1 (0.6-1.4)	0.8 (0.7-1.3)	1.0 (0.7-1.3)	1.1 (1.0-1.7)	1.2 (0.9-1.9)
TC	240 (195-299)	271 (205-307)	249 (202-340)	252 (208-279)	248 (194-298)	272 (211-322)
TG	177 (88-474)	161 (79-330)	125 (54-295)	129 (62-200)	182 (67-240)	219 (101-293)

Lipids and lipoproteins are presented in mg/dL.

Lipid Profile Two Weeks After Treatment

Characteristic (Median %Δ)	Placebo	50 mg/wk	100 mg/wk	200 mg/wk	300 mg/wk	400 * mg/wk
ApoB	7 %	-22 % P=0.07	-23 % P=0.001	-47 % P=0.0002	-61 % P=0.0003	Data Pending*
LDL-C	2 %	-12 % P=0.33	-22 % P=0.01	-42 % P=0.0002	-62 % P=0.0003	
Non-HDL-C	4 %	-17 % P=0.25	-21 % P=0.02	-44 % P=0.0002	-54 % P=0.0003	
VLDL-C	-17 %	-14 % P=0.43	-14 % P= 0.93	-54 % P=0.006	-52 % P=0.04	
HDL-C	2 %	9 % P=0.28	5 % P=0.40	-1 % P=0.72	-15 %† P=0.009	
Apo A1	0%	8 % P=0.01	5 % P=0.19	-2 % P=0.96	-14 % P=0.06	
Apo B:A1	11%	-22 % P=0.007	-26 % P=0.002	-46 % P=0.0002	-51 % P=0.0003	
TC	6 %	-12 % P=0.33	-15 % P=0.09	-34 % P=0.0002	-46 % P=0.0003	
TG	-13 %	-7 % P=0.65	-22 % P=0.54	-46 % P=0.02	-43 % P=0.04	

*Data Pending – Will be included on the poster at time of meeting.

†HDL-C measurements for this study were performed using a direct method that can be inaccurate when LDL-C levels are low. Parallel evaluation of the 300 mg/wk cohort in this study using a common precipitation method showed an 11% increase in HDL-C.

Liver Transaminase

ALT Elevations*	Placebo (n = 8)**	50 mg (n = 8)	100 mg (n = 8)	200 mg (n =8)	300 mg† (n = 8)	400 mg‡ (n = 8)
Dosing	0	0	0	0	0	
Follow-up	0	0	0	1		

* ALT elevation ≥ 3xULN on two consecutive measures at least 7 days apart.
** 2 subjects not included. †Cohort in f/u period. ‡Cohort in dosing period.

ApoB*

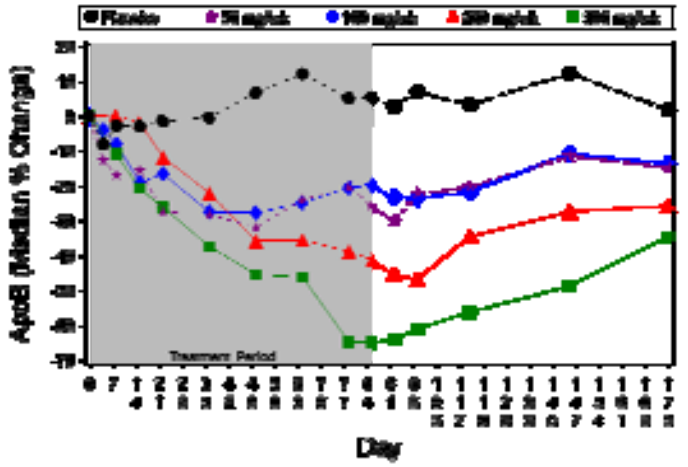
Treatment Period
Day

LDL-C*

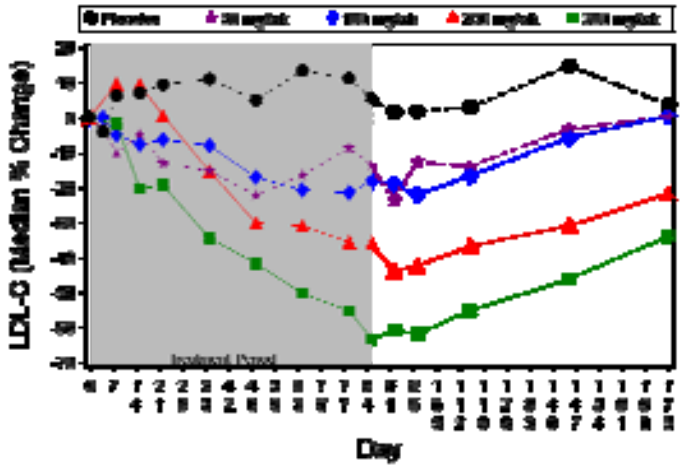
Day

TG*

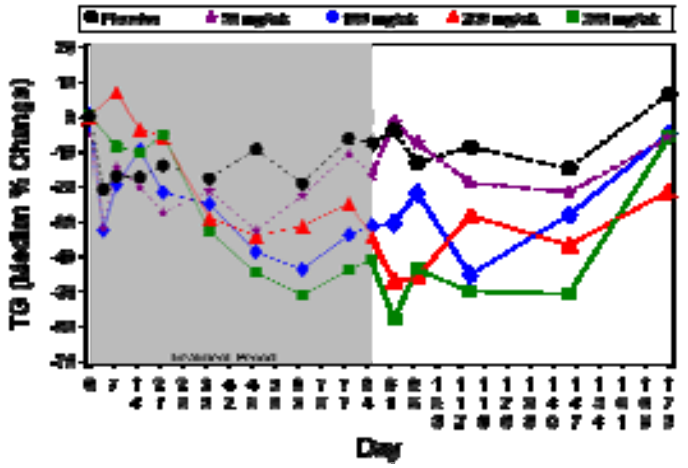
Day

* Data Pending – 400 mg/wk data will be included on poster at time of meeting.

Safety Observations

- One SAE (meningoencephalitis) occurred that was considered unrelated to drug.
- The most common adverse event was mild, painless erythema at site of injection.
- One subject had consecutive ALT elevations ≥ 3x ULN during follow-up, with no elevation in total bilirubin.
- Recently dosed cohorts continue in their 6-month post-dosing follow-up period.

Summary of Results

- Potent, dose-dependent reductions in atherogenic lipids and lipoproteins with maximum XX% (P=xxx) and YY% (P=xxx) reduction in apoB and LDL-C at 400 mg/wk respectively.
- Prolonged effects consistent with > 30-day terminal elimination half-life.
- Clinically significant reductions in TG and VLDL-C levels with maximum XX% (P=xxx) and YY% (P=xxx) reductions at 400 mg/wk respectively.

Conclusions

- Reductions in apoB-containing lipoproteins exceed those reported for statins in other studies
- Clinically significant reductions in triglycerides
- No unexpected toxicities
- Further studies are required to determine the generalizability of these results.

References

1. Libby P. JACC 2005;46:1225-28.
2. Cannon CP. JAMA 2005;294:2492-94.
3. Crooke ST. Annu Rev Med 2004;55:61-95.
4. Kastelein JJP, et al. Circulation 2006;114:1729-35.
5. Kastelein JJP. In Clinical Trials of Lipid-Lowering Therapy, 56th Annual Scientific Sessions 2007, ACC, New Orleans, LA March 24-27.